EFFICACY OF PUR1900, AN INHALED ANTIFUNGAL THERAPY, IN A GUINEA PIG MODEL OF INVASIVE PULMONARY ASPERGILLOSIS

A.K. Curran, J.M. Perry, and D.L. Hava

Pulmatrix, Inc., Lexington, Massachusetts, USA

pulmatrix

INTRODUCTION

Pulmonary fungal infections in asthma and cystic fibrosis patients range from allergic bronchopulmonary aspergillosis (ABPA) to invasive aspergillosis, are underdiagnosed[1] and can be a significant source of morbidity and mortality. Oral triazole treatment is commonly prescribed, yet historically, oral bioavailability and achieved lung concentrations are variable and often subtherapeutic[2]. In addition, triazoles like itraconazole have multiple drug-drug interactions (DDIs) that limit their utility as oral drugs. Pulmatrix has developed Pulmazole™ (PUR1900), an inhaled dry powder formulation of itraconazole that is formulated using our proprietary dry powder platform iSPERSE[3], engineered to a mass median aerosol diameters (MMAD) of ~3μm and high fine particle dose (FPD; % of the nominal dose < 5μm), which results in more than 50% of the nominal dose reaching the lungs with reduced throat deposition. Notably, the aerosol target range of PUR1900 is similar to that of Aspergillus conidia, which, in theory, should result in PUR1900 delivery to lung sites where aspergillus spores also deposit.

AIMS

- To assess efficacy of PUR1900 (Pulmazole™) in a model of invasive aspergillosis
- To compare efficacy against Sporanox®
- To assess systemic and lung exposure with both dose routes

METHODS

Both *A. fumigatus challenge* and PUR1900 dose were delivered using a directed flow inhalation system as shown in Fig 1.

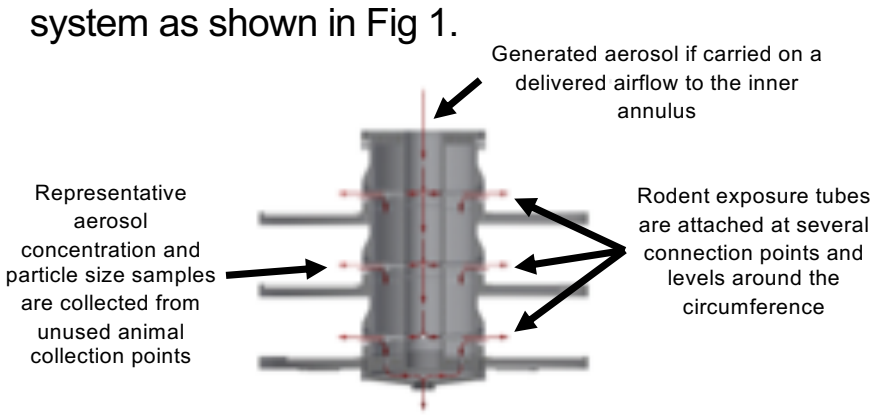
Generated aerosol if carried on a delivered airflow to the inner annulus
Representative aerosol concentration and particle size samples are collected from unused animal collection points
Rodent exposure tubes are attached at several connection points and levels around the circumference

Figure 1, Inhalation Dosing. Aerosol is generated as required and delivered *via* a central annulus from which each animal, restrained in a restraint tube, receives individual and identical aerosol exposure. Exhaust is collected from the tubes and removed using the outer annulus. This mechanism allows for a consistent delivery of aerosol with no filtering or rebreathing by animals at different points on the system

Pathogen Challenge

All manipulations using *A. fumigatus* conidia were performed at room temperature conducted using BSL-2/ABSL-2 containment facilities and practices. Potato dextrose agar (PDA) plates were inoculated with *A. fumigatus* and incubated at 37±2°C for 7 days. Conidia were harvested and processed, resulting in a dark green suspension that was stored on wet ice until use. All guinea pigs were immunosuppressed with cyclophosphamide and cortisone acetate on two occasions and challenged *via* nose-only inhalation for 45 minutes using a nebulizer suspension at a target concentration of approximately 5×10^8 conidia/mL.

Inhalation dosing

Eight animals per group were assigned to one of 4 dose groups as detailed in the table.

Delivered and Achieved Doses

Group	Treatment	Target Dose (mg/kg/day)	Achieved Dose (mg/kg/day)	MMAD (μm)
1	Placebo	0	0	2.25
2	PUR1900	10	10.1	2.62
3	PUR1900	30	26.9	
4	Sporanox	10	10	-

Pulmonary deposited dose is considered to be 10% of the achieved dose in small animal species[4].

Treatment aerosols were generated with a rotating brush generator (RBG) into a directed flow nose-only inhalation system. Aerosol concentration was measured daily and particle size was measured on at least one occasion per dose level. Animals were treated daily for 10 days, starting one day after infection and monitored for an additional 4 days after the end of dosing.

Lung and systemic itraconazole levels were assessed from 3 animals per group immediately after dose and 24h after dose on Day 1 and from any surviving animals on Day 14. In addition, body temperature, body weight and survival were monitored throughout the study and pulmonary fungal burden was assessed from any surviving animals on Day 14. Statistical analysis was performed using Student's t-test for paired comparisons and Log-rank (Mantel-Cox) Test for survival (Prism 5.0, GraphPad Software, Inc.).

RESULTS

Bioanalysis

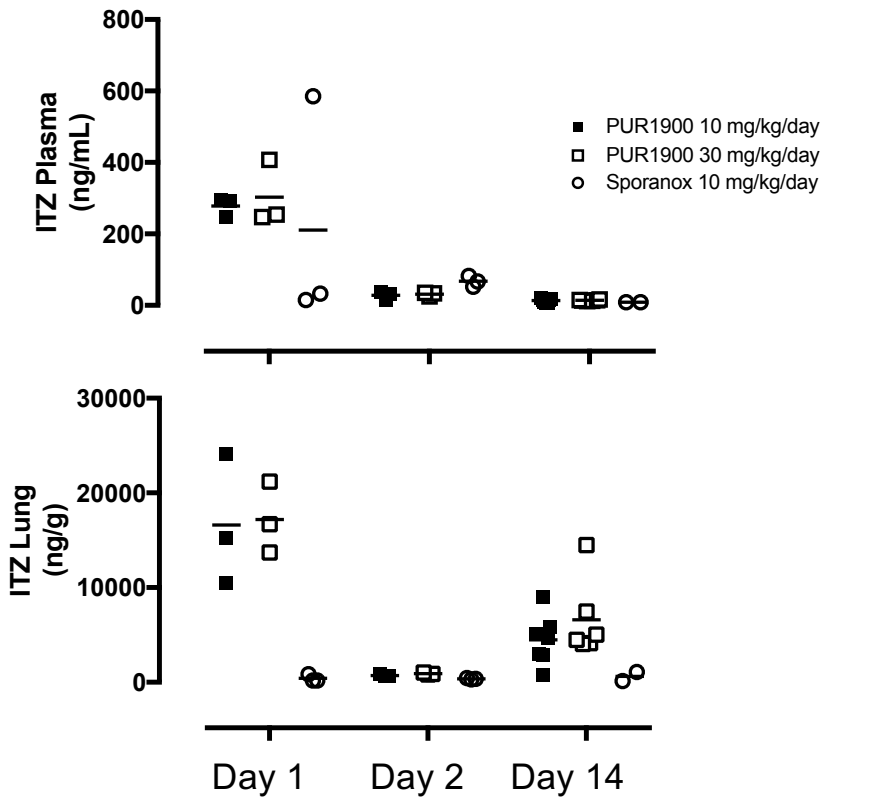
ITZ Plasma (ng/mL)
0
200
400
600
800
PUR1900 10 mg/kg/day
PUR1900 30 mg/kg/day
Sporanox 10 mg/kg/day
ITZ Lung (ng/g)
0
10000
20000
30000
Day 1
Day 2
Day 14

Fig 2, Bioanalysis. On Day 1, immediately after dose, plasma itraconazole levels were generally low for both dose routes and very variable after oral Sporanox. Lung levels were high after inhalation dosing but not after oral dose. By 24 hours after dose, both plasma and lung levels had reduced significantly. By Day 14 (4 days after the last dose), significant lung levels remained in the inhalation dosed animals only, at levels that would be considered sufficient to provide efficacy, whereas only trace lung levels were seen in orally dosed animals. Plasma exposure in either inhaled or orally dosed animals was low for all groups on Day 14.

Body Weight and Temperature

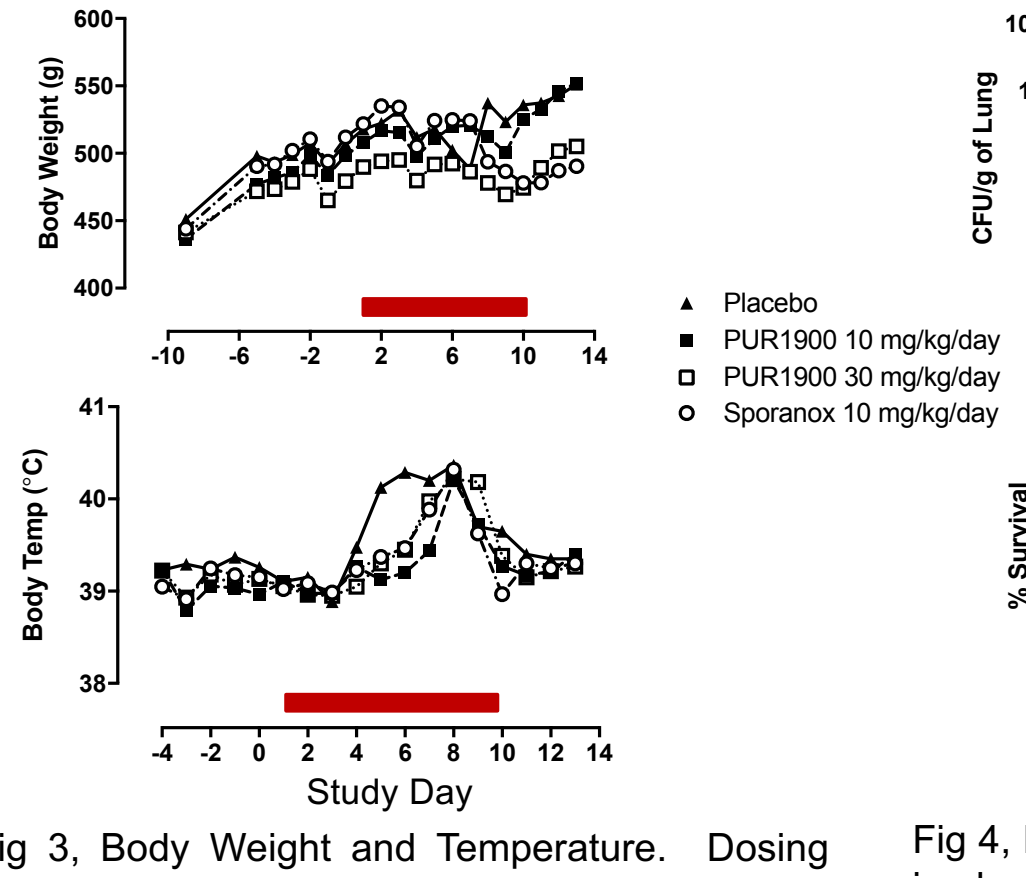
Body Weight (g)
400
450
500
550
600
-10
-6
-2
2
6
10
14
Placebo
PUR1900 10 mg/kg/day
PUR1900 30 mg/kg/day
Sporanox 10 mg/kg/day
Body Temp (°C)
38
39
40
41
-4
-2
0
2
4
6
8
10
12
14
Study Day

Fig 3, Body Weight and Temperature. Dosing period is shown as a red bar. Exposure to *A. fumigatus* resulted in weight loss between Days 4 and 9 in all groups. There was no significant effect of any treatment on body weight change. Similarly, exposure to *A. fumigatus* resulted in an increase in body temperature after Day 4. Treatment with either PUR1900 or Sporanox failed to significantly effect the magnitude of the temperature response but did delay the onset slightly. The reduction in temperature ion control animals is an artifact secondary to the death of infected animals.

Fungal Burden and Survival

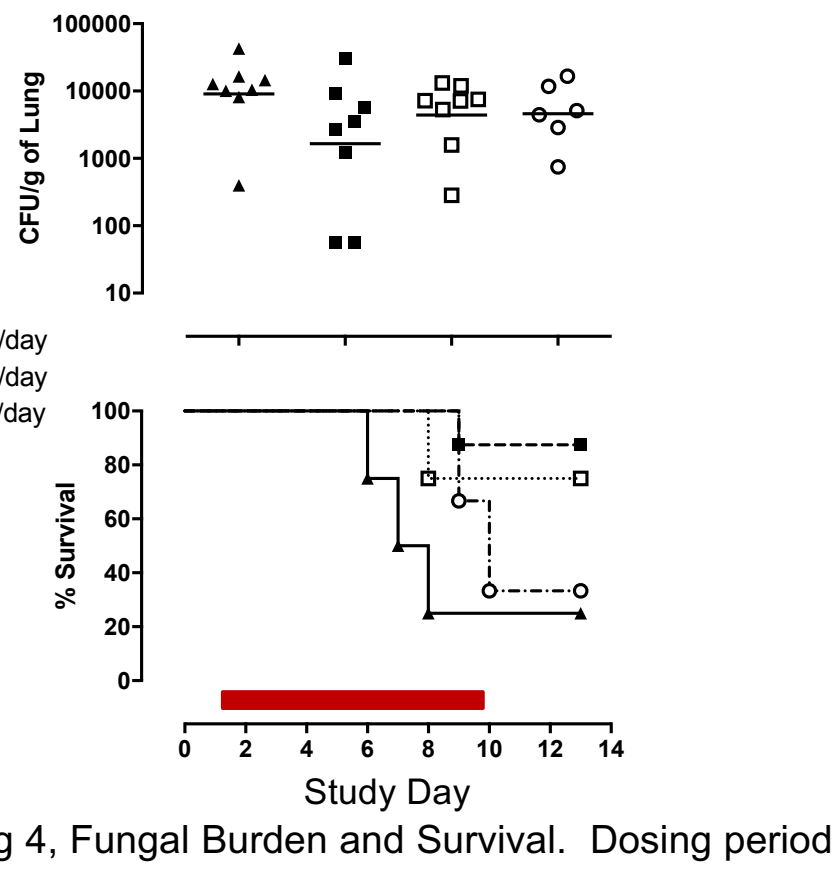
CFU/g of Lung
10
100
1000
10000
100000
% Survival
0
20
40
60
80
100
0
2
4
6
8
10
12
14
Study Day

Fig 4, Fungal Burden and Survival. Dosing period is shown as a red bar. On day of euthanasia or Day 14, 4 days after the last dose, lung samples were collected for quantitative fungal burden. There was no significant difference between placebo and test article treated animals, though there was a tendency for lower burden in treated animals. Survival was significantly increased in PUR1900-treated animals relative to both placebo control and Sporanox, both in terms of numbers of animals surviving and average duration of survival, indicating a significant advantage of inhaled dosing versus oral.

CONCLUSIONS

- Inhaled PUR1900 provides higher lung exposure than oral Sporanox, relatively low systemic exposure and a high lung:plasma ratio.
- These data indicate that Pulmazole™ (PUR1900) shows potential as an inhaled therapy for pulmonary fungal infection.

REFERENCES

1. Armstead J, Morris J, Denning DW (2014) Multi-Country Estimate of Different Manifestations of Aspergillosis in Cystic Fibrosis. PLoS ONE 9(6): e98502
2. Sermet-Gaudelus, et al. (2001) Sputum Itraconazole Concentrations in Cystic Fibrosis Patients. Antimicrob. Agents Chemother. 45(6):1937
3. Sung JC, et. al. (2011) iSPERSE: Formulation and In Vitro Characterization of a Novel Dry Powder Drug Delivery Technology. RDD Europe
4. Wolff R.K., M.A. Dorato (1993). Toxicologic Testing of Inhaled Pharmaceutical Aerosols. Critical Reviews in Toxicology. 23(4): 343-369